Combination of RAS(ON) Mutant-Selective and Multi-Selective Inhibitors Sensitizes Immune-Refractory, RAS-Driven Preclinical Models to Immunotherapy

Mariela Moreno Ayala, Cristina Blaj, Marie Menard, Avery Salmon, Jasmine Lee, Ximo Pechuan-Jorge, Rashi Raghulan, Swetha Ganesh, Anna Pham, Vivian Morton, Miguel Sandoval, Jacqueline AM Smith, Elsa Quintana

Revolution Medicines, Redwood City CA USA

Introduction

- Oncogenic RAS promotes carcinogenesis by sustaining cell proliferation and orchestrating immune escape
- We and others have shown that RAS inhibition combined with immune checkpoint blockade (ICB) prolongs durability of response in immunogenic, RAS-driven preclinical models[1,2]
- Clinical data have shown that most patients treated with KRAS G12C(OFF) inhibitors develop resistance through reactivation of the RAS pathway
- We have previously demonstrated that the RAS(ON) doublet combinate ion of a RAS(ON) G12C-selective inhibitor with a RAS(ON) multi-selective inhibitor can overcome mechanisms of intrinsic and acquired resistance in an immune-refractory preclinical NSCLC model[1]
- Here we evaluated if the RAS(ON) doublet combination of elironrasib (RMC-6291) or zoldonrasib (RMC-9805), a RAS(ON) G12C-selective inhibitor and a RAS(ON) G12D-selective inhibitor, respectively, with the RAS(ON) multi-selective inhibitor daraxonrasib (RMC-6236) can sensitize traditionally hard to treat, immune-refractory preclinical models of RAS mutant NSCLC and PDAC to anti-PD-1

Revolution Medicines' Tri-Complex Inhibitor Platform to Target Oncogenic RAS

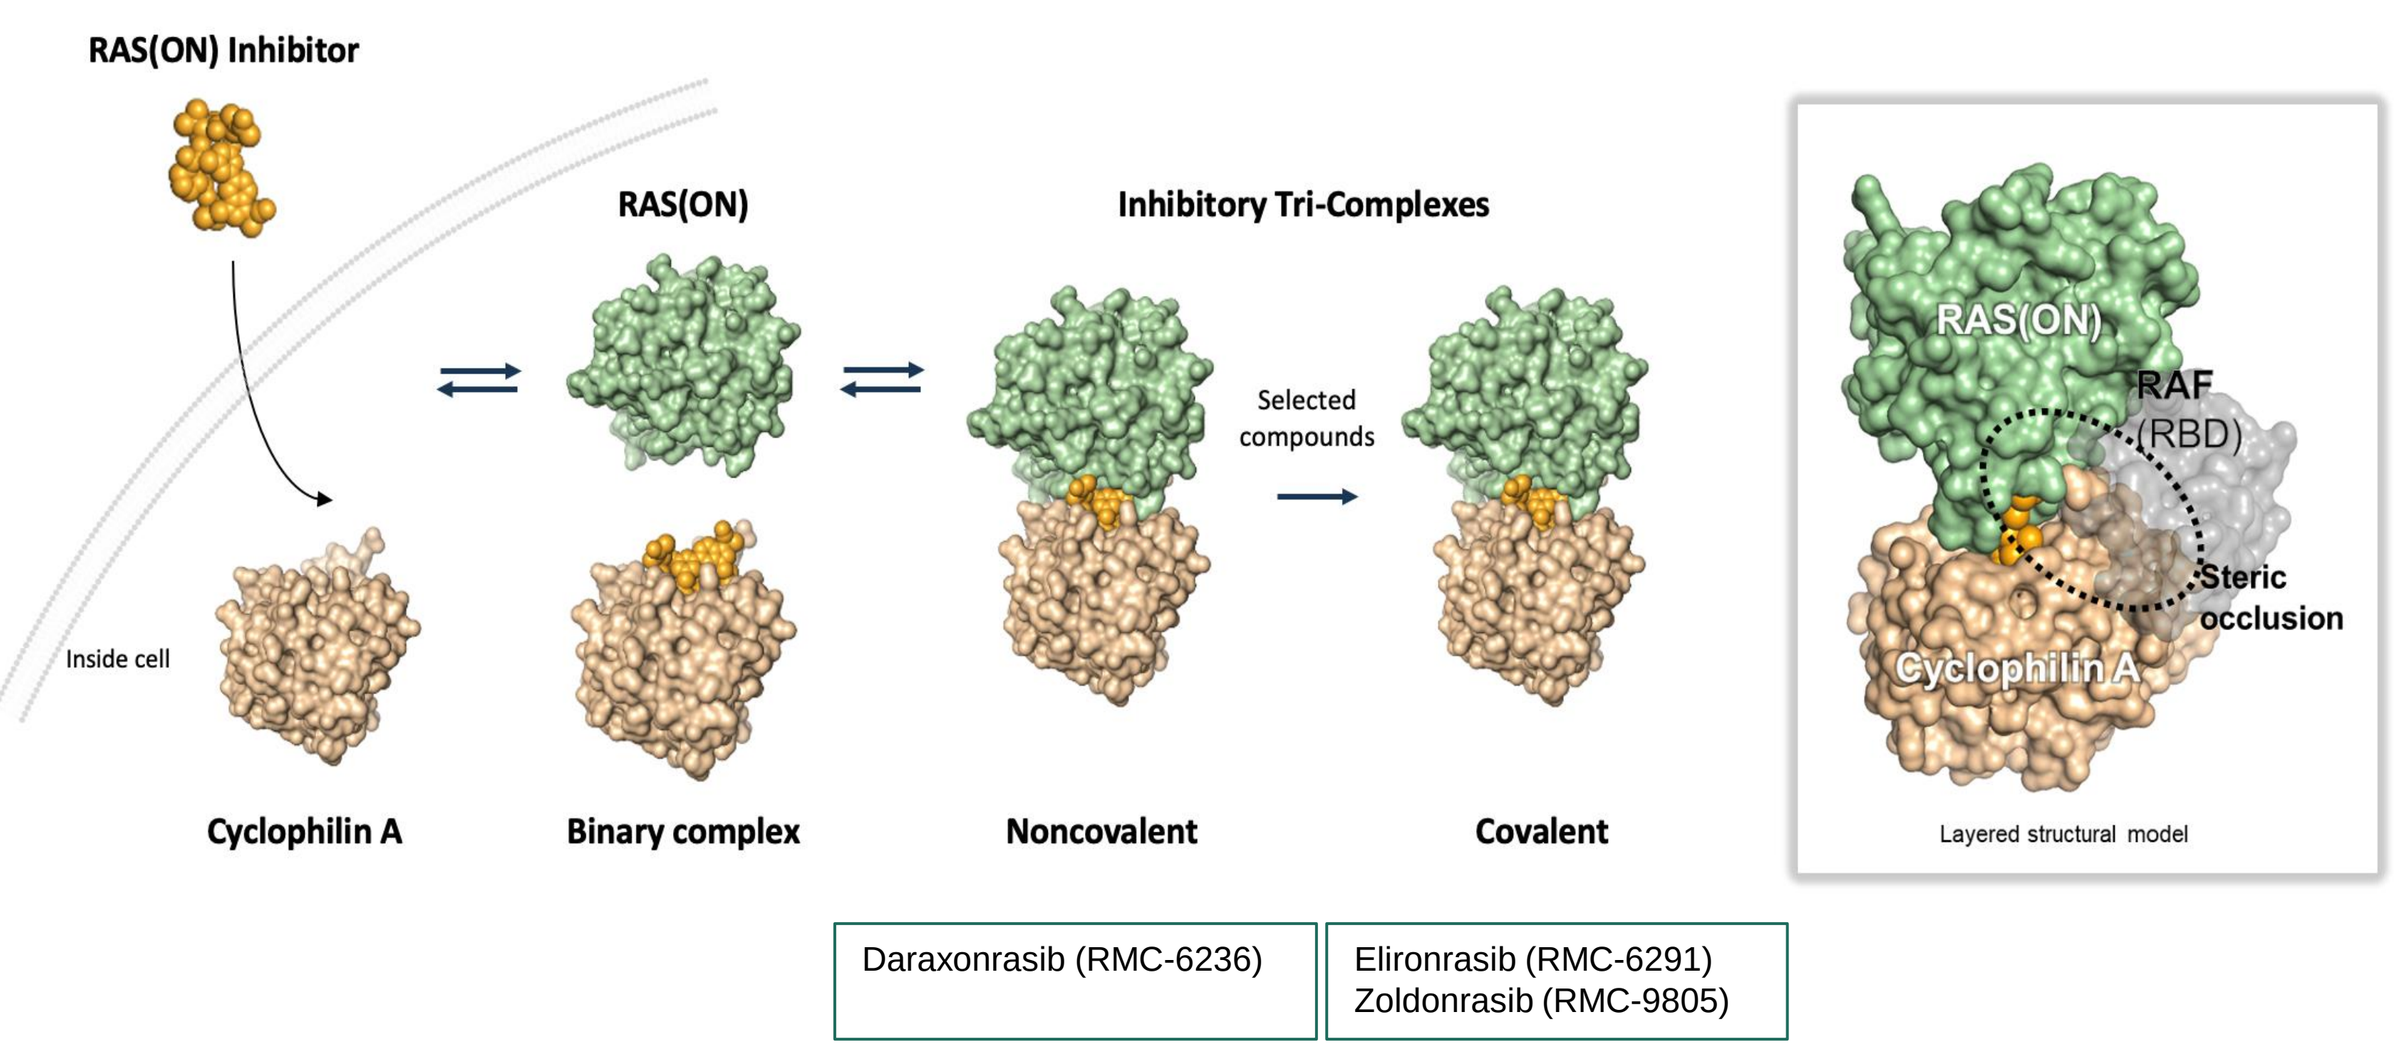
RAS(ON) Inhibitor
RAS(ON)
Inhibitory Tri-Complexes
Selected compounds
Inside cell
Cyclophilin A
Binary complex
Noncovalent
Covalent
RAS(ON)
RAF (RBD)
Steric occlusion
Cyclophilin A
Layered structural model
Daraxonrasib (RMC-6236)
Elironrasib (RMC-6291)
Zoldonrasib (RMC-9805)

Preclinical strategy to test the combination of RAS(ON) doublet + CPI in immuno-refractory tumor models

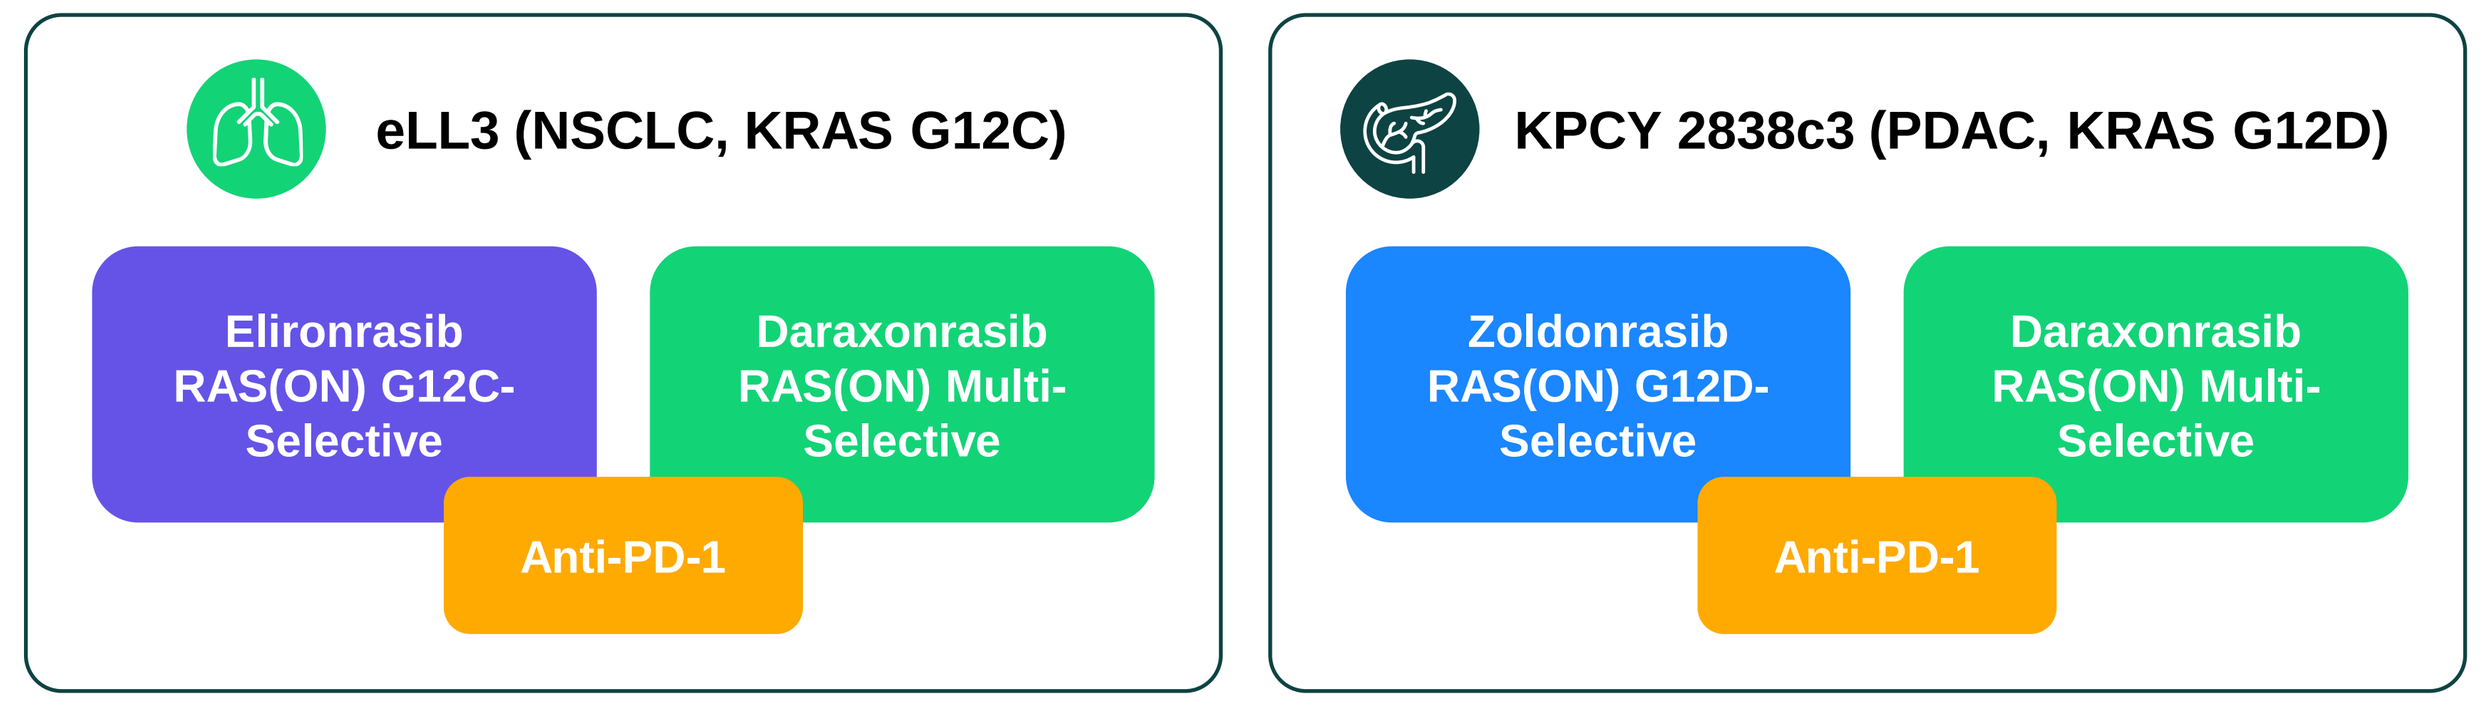
eLL3 (NSCLC, KRAS G12C)
Elironrasib RAS(ON) G12C-Selective
Daraxonrasib RAS(ON) Multi-Selective
Anti-PD-1
KPCY 2838c3 (PDAC, KRAS G12D)
Zoldonrasib RAS(ON) G12D-Selective
Daraxonrasib RAS(ON) Multi-Selective
Anti-PD-1

Key Results

Elironrasib plus daraxonrasib RAS(ON) doublet maximizes RAS pathway suppression and sensitizes an immune refractory NSCLC model to anti-PD-1

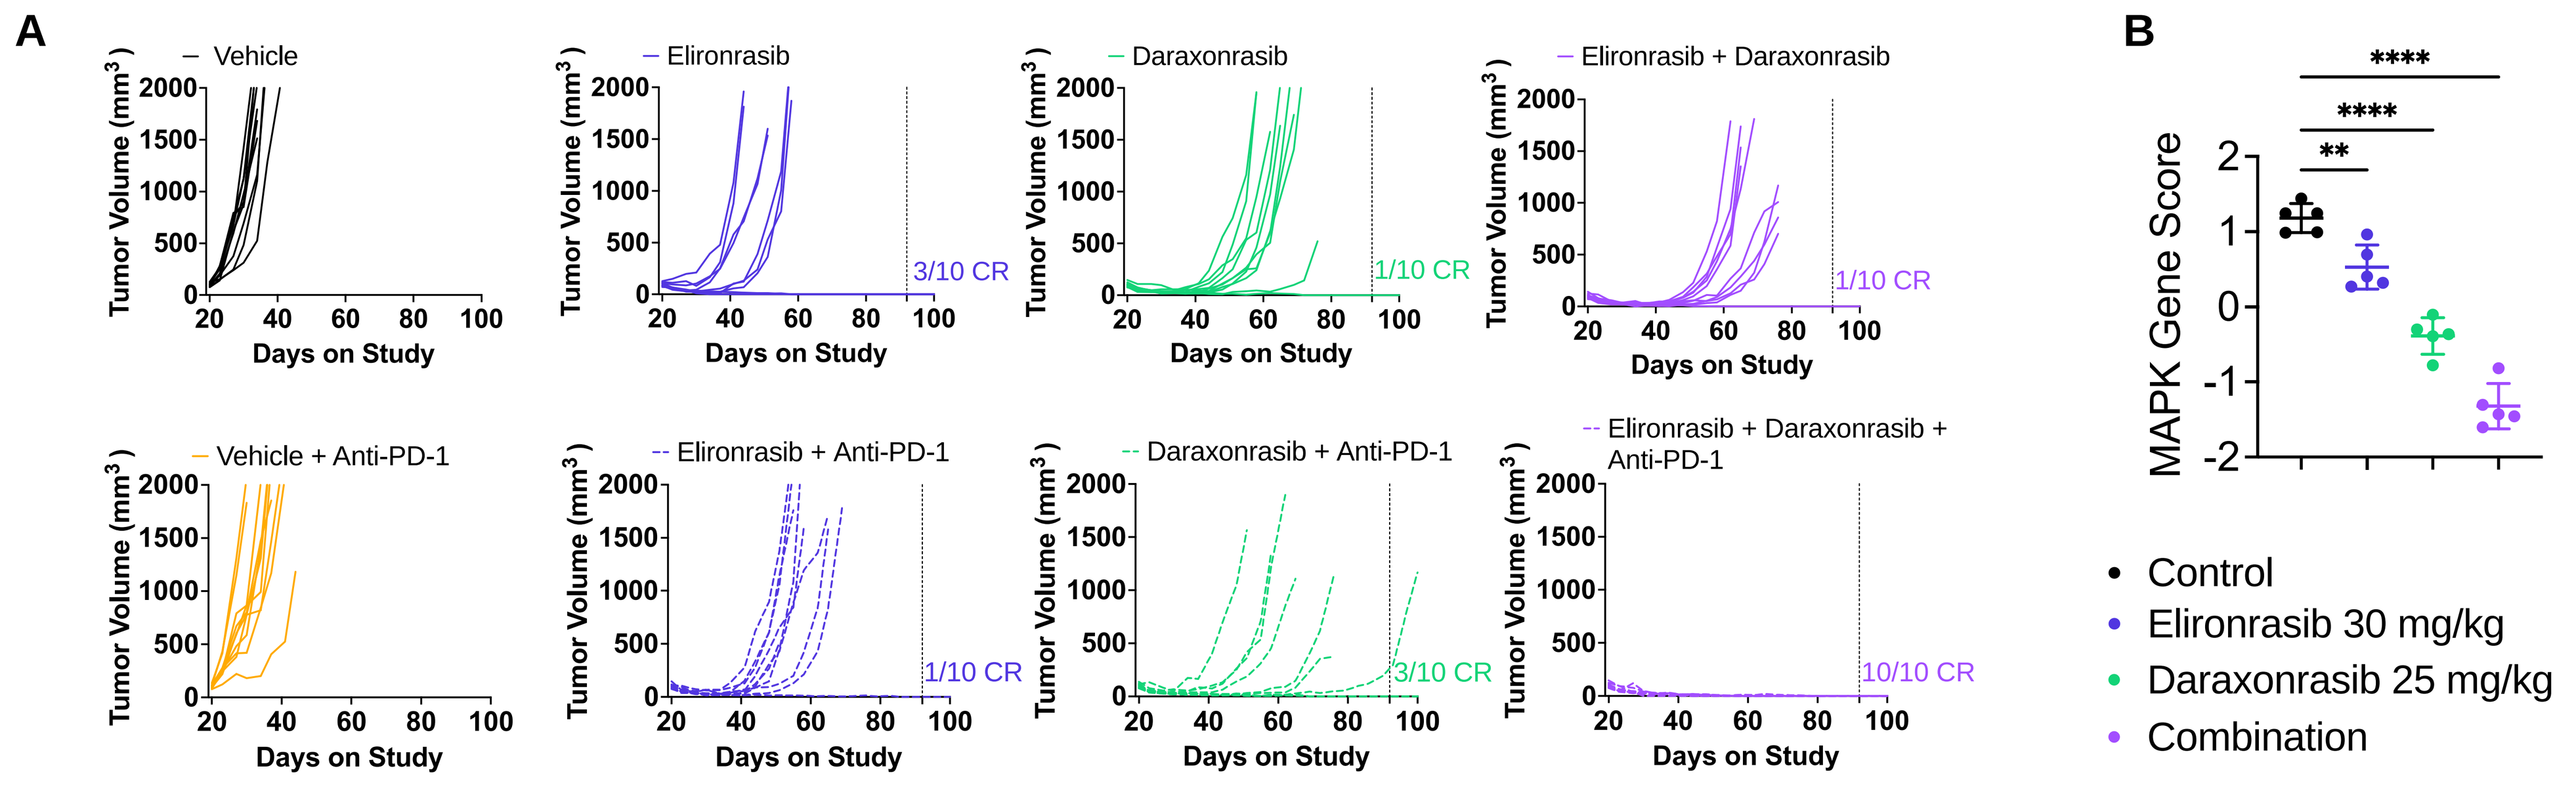
A
Vehicle
Elironrasib
Daraxonrasib
Elironrasib + Daraxonrasib
Vehicle + Anti-PD-1
Elironrasib + Anti-PD-1
Daraxonrasib + Anti-PD-1
Elironrasib + Daraxonrasib + Anti-PD-1
Tumor Volume (mm³)
Days on Study
3/10 CR
1/10 CR
1/10 CR
1/10 CR
3/10 CR
10/10 CR
B
MAPK Gene Score
Control
Elironrasib 30 mg/kg
Daraxonrasib 25 mg/kg
Combination

- All mice with complete regressions established immunological memory and rejected the tumors upon rechallenge

A. 3LL ΔNRAS tumors treated with elironrasib at 30 mg/kg po qd, daraxonrasib at 25 mg/kg po qd and anti-PD-1 at 10 mg/kg ip biw; dotted line indicates treatment stop; days on study represent days post tumor inoculation B. PROGENy score of bulk RNAseq data from 3LL ΔNRAS tumors treated with elironrasib at 30 mg/kg po qd, daraxonrasib at 25 mg/kg po qd or the combination at 24 hours post 7 days of treatment.

Zoldonrasib plus daraxonrasib RAS(ON) doublet maximizes RAS pathway suppression and sensitizes an immune-refractory GEM-derived PDAC model to anti-PD-1

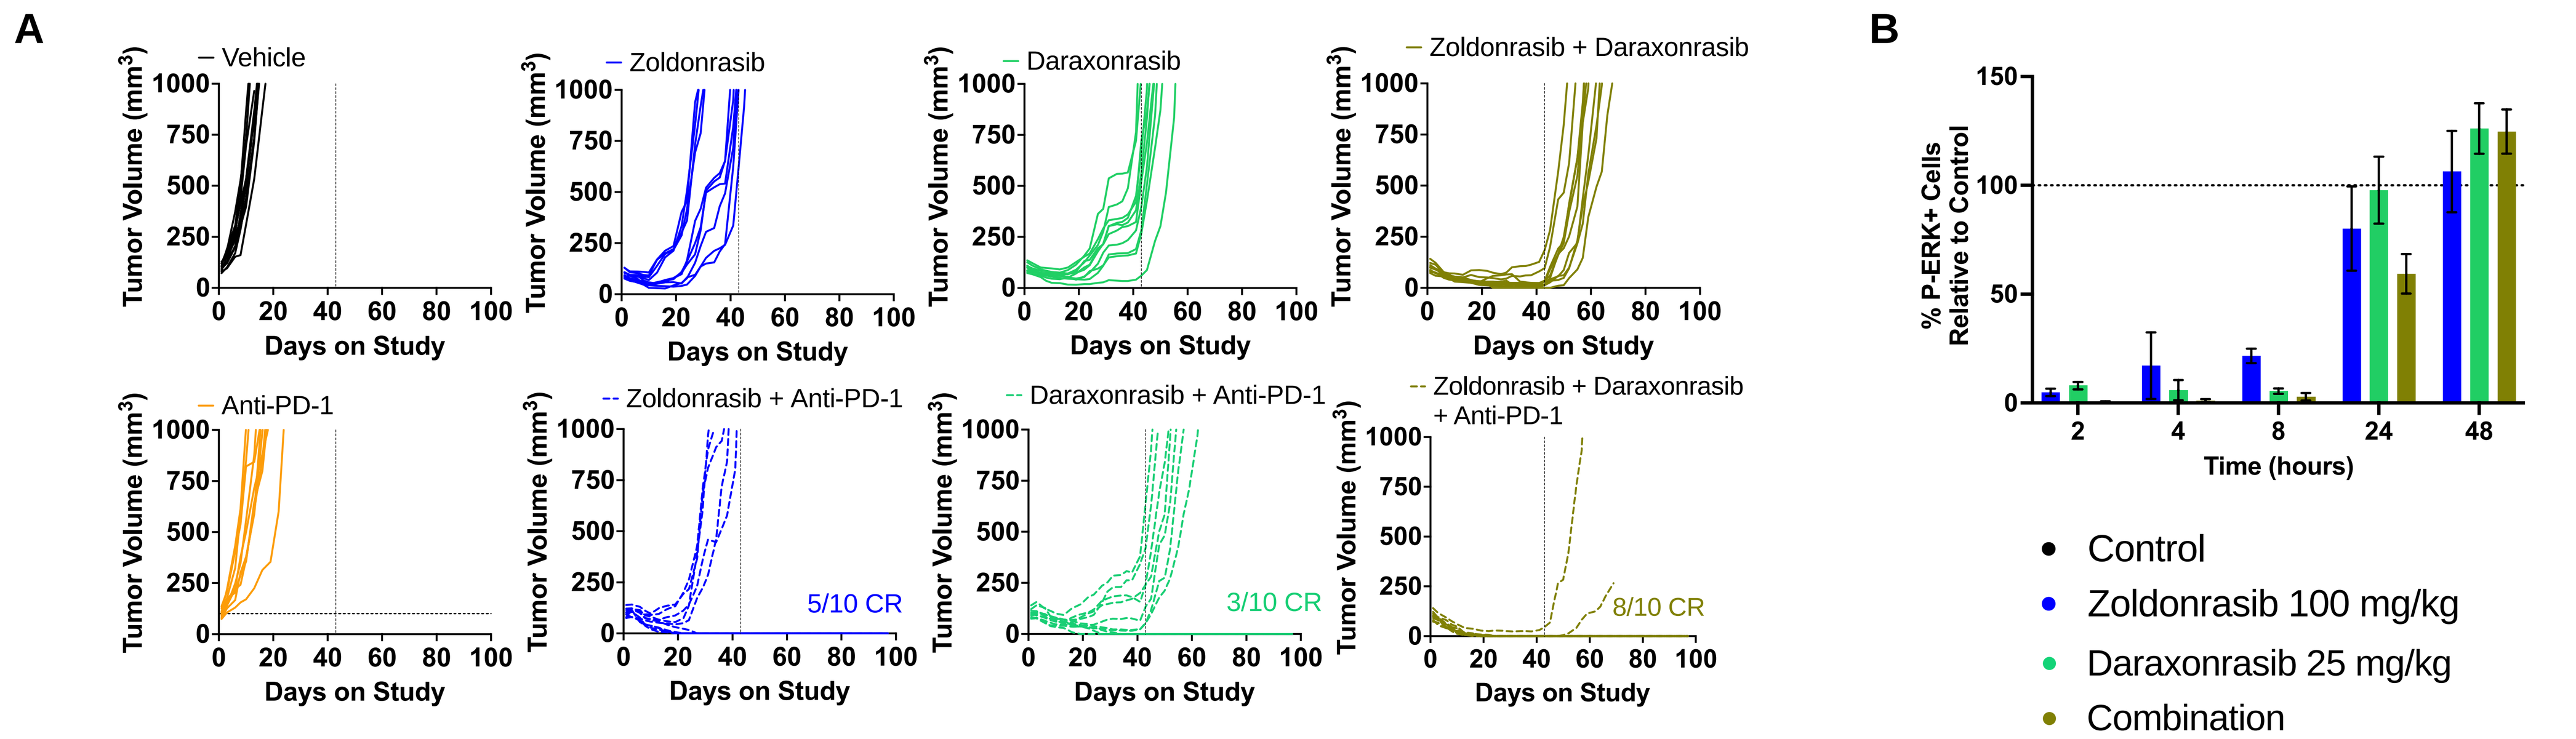
A
Vehicle
Zoldonrasib
Daraxonrasib
Zoldonrasib + Daraxonrasib
Anti-PD-1
Zoldonrasib + Anti-PD-1
Daraxonrasib + Anti-PD-1
Zoldonrasib + Daraxonrasib + Anti-PD-1
Tumor Volume (mm³)
Days on Study
5/10 CR
3/10 CR
8/10 CR
B
% P-ERK+ Cells Relative to Control
Time (hours)
Control
Zoldonrasib 100 mg/kg
Daraxonrasib 25 mg/kg
Combination

- Zoldonrasib and Daraxonrasib as single agents achieve combination benefit with anti-PD-1

KPCY 2838c3 tumors treated with zoldonrasib at 100 mg/kg po qd, daraxonrasib at 25 mg/kg po qd and anti-PD-1 at 10 mg/kg ip biw; dotted line indicates treatment stop for the RAS(ON) inhibitors; days on study represent days on treatment. B. IHC quantification of p-ERK N = 3 mice/group of tumors treated with zoldonrasib at 100 mg/kg po qd, daraxonrasib at 25 mg/kg po qd or the combination at indicated time points.

Elironrasib and daraxonrasib favorably modulate the tumor microenvironment, and the combination additionally increases antigen presentation in a NSCLC model

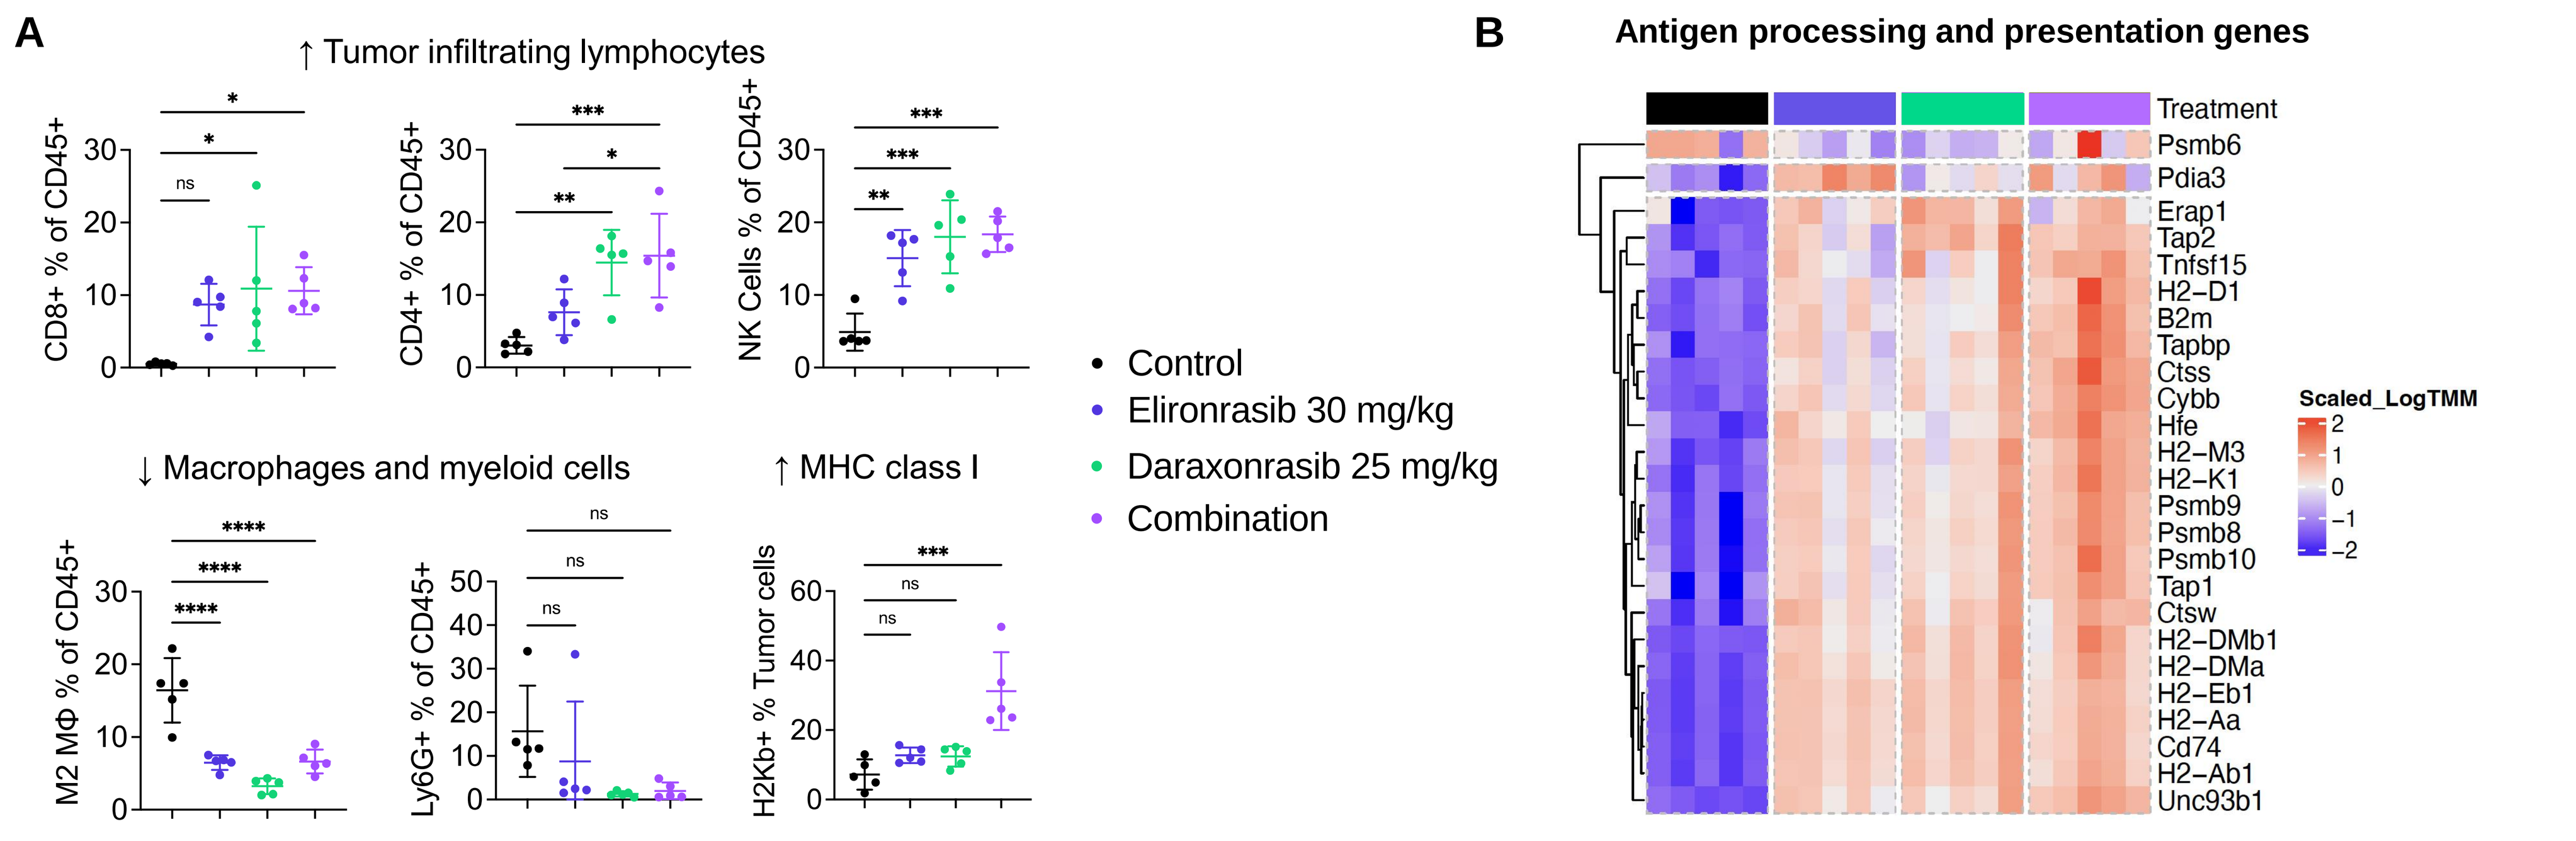
A
↑ Tumor infiltrating lymphocytes
CD8+ % of CD45+
CD4+ % of CD45+
NK Cells % of CD45+
↓ Macrophages and myeloid cells
M2 MΦ % of CD45+
Ly6G+ % of CD45+
↑ MHC class I
H2Kb+ % Tumor cells
Control
Elironrasib 30 mg/kg
Daraxonrasib 25 mg/kg
Combination
B
Antigen processing and presentation genes
Treatment
Scaled_LogTMM

A. Flow cytometric analysis and B. Bulk RNAseq analysis of 3LL ΔNRAS tumors treated with elironrasib at 30 mg/kg po qd, daraxonrasib at 25 mg/kg po qd or the combination at 24 hours post 7 days of treatment. N = 5 mice/group. One-way ANOVA (*P<0.05, **P <0.01, ***P <0.001, ****P<0.0001).

The combination of zoldonrasib and daraxonrasib achieves favorable transformation of the tumor microenvironment and increase in antigen presentation in a PDAC model

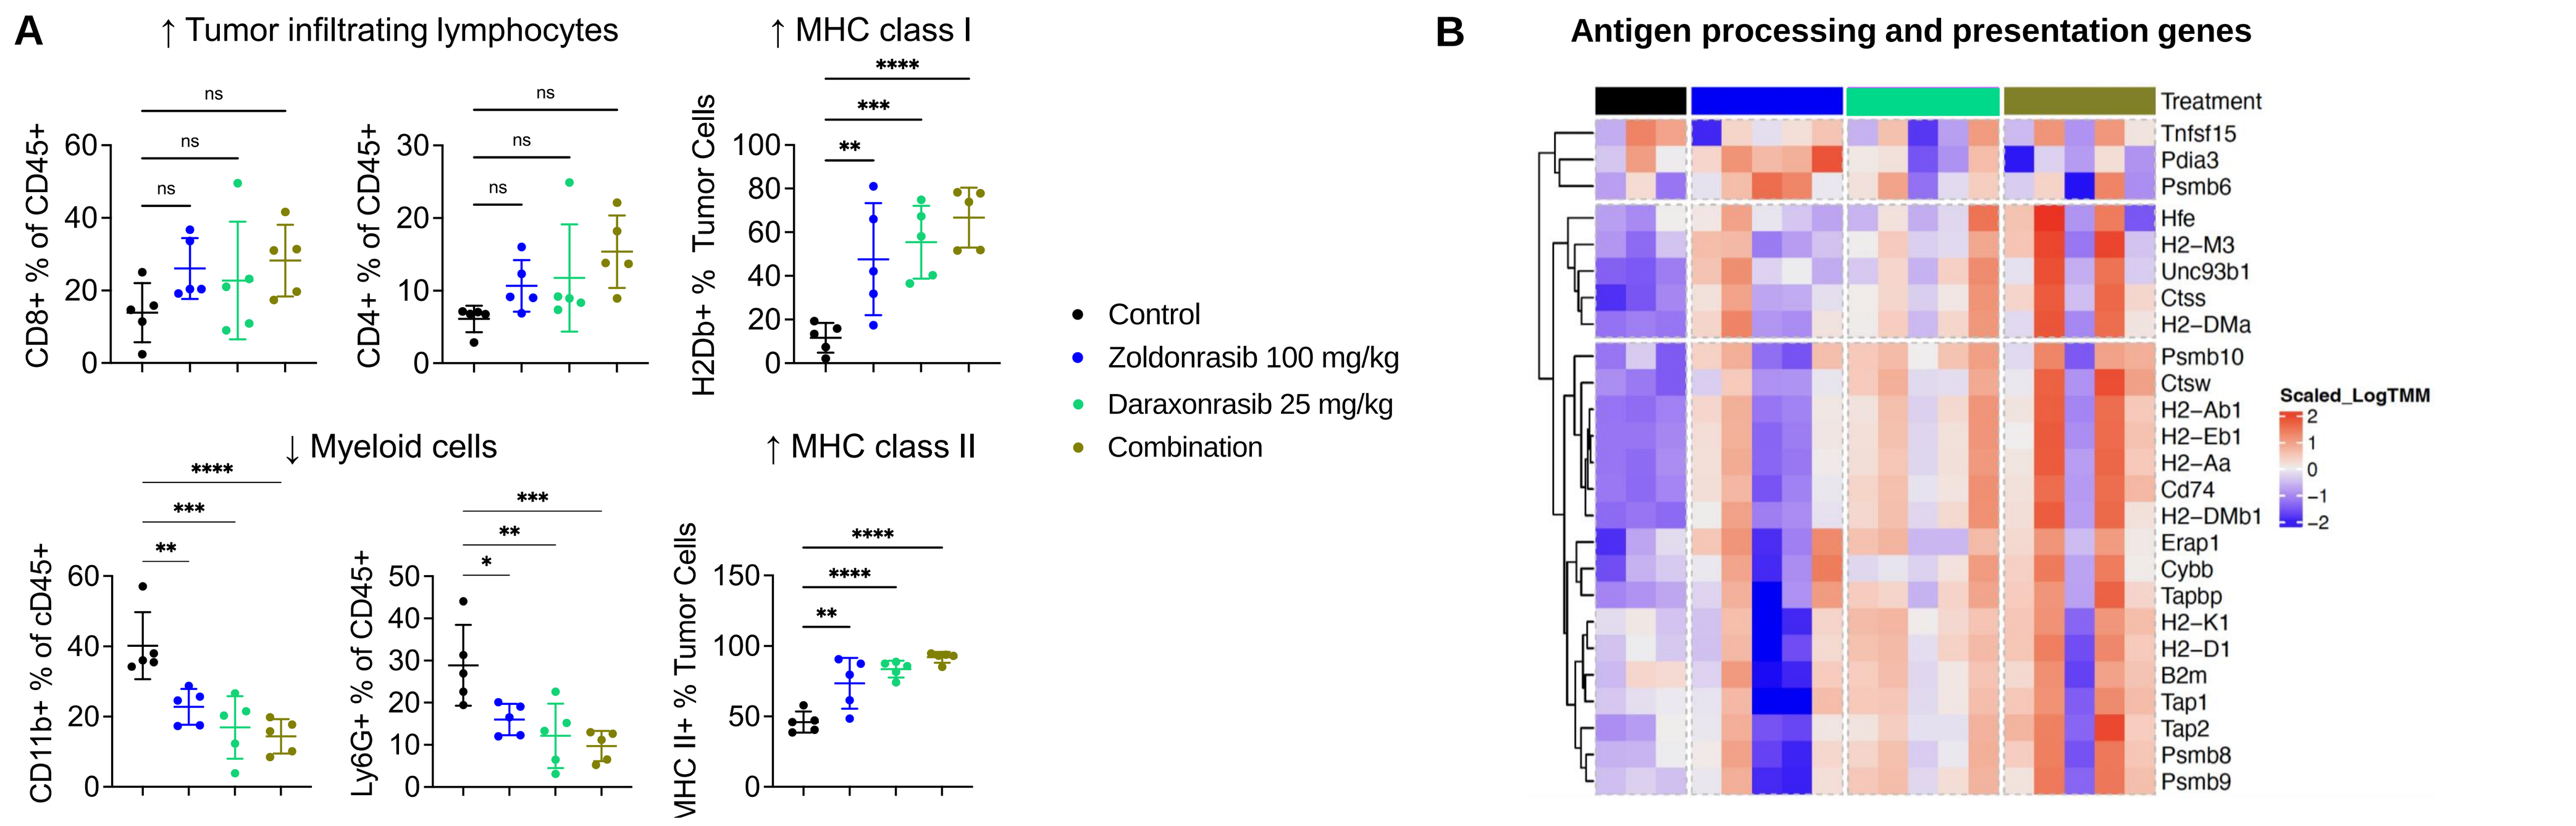
A
↑ Tumor infiltrating lymphocytes
CD8+ % of CD45+
CD4+ % of CD45+
↑ MHC class I
H2Db+ % Tumor Cells
↓ Myeloid cells
CD11b+ % of cD45+
Ly6G+ % of CD45+
↑ MHC class II
MHC II+ % Tumor Cells
Control
Zoldonrasib 100 mg/kg
Daraxonrasib 25 mg/kg
Combination
B
Antigen processing and presentation genes
Treatment
Scaled_LogTMM

A. Flow cytometric analysis and B. Bulk RNAseq analysis of tumors treated with zoldonrasib at 100 mg/kg po qd, daraxonrasib at 25 mg/kg po qd or the combination at 24 hours post 7 days of treatment at 24 hours post 8 days of treatment. N = 5 mice/group. One-way ANOVA (*P<0.05, **P<0.01, ***P<0.001, ****P<0.0001).

Conclusion

- RAS(ON) doublet combination of RAS(ON) mutant-selective and multi-selective inhibitors achieved deeper and more durable pathway suppression and prolonged durability of response in preclinical models of RAS mutant NSCLC and PDAC
- The RAS(ON) doublet reversed the immune-evasion mechanisms governed by oncogenic RAS, decreasing myeloid derived suppressor cells and favoring the infiltration with T cells
- The RAS(ON) doublet upregulated MHC class I expression, increasing the probability of antigen presentation and recognition of the tumor by T cells and sensitized immune-refractory tumors to ICB to induce durable CRs and immunological memory
- Collectively, these preclinical data support the clinical evaluation of the RAS(ON) doublets in combination with ICB in patients with RAS-driven cancers
- Clinical evaluation of RAS(ON) inhibitors with pembrolizumab is ongoing (NCT06162221)

Abbreviations:
ANOVA, analysis of variance; anti-PD-1, anti-programmed cell death protein 1; biw, biweekly; CRs, complete regressions; CPI, checkpoint inhibitor; ENA, European Network of Addiction; GEM, genetically engineered mouse; ICB, immune checkpoint blockade; IHC, immunohistochemistry; KRAS, Kirsten rat sarcoma virus oncogene; MHC, major histocompatibility complex; NSCLC, non-small cell lung cancer; PDAC, pancreatic ductal adenocarcinoma; PROGENy, pathway respiration and organo-genic influenced network lysing network phenotype; RAS, rat sarcoma virus oncogene; RNAseq, rna sequencing

References:
1. Blaj et al, ENA, [Poster Abstract #PB044] (2022)
2. Anastasiou et al, Nature Communications 15(1):8146 (2024)

AACR
April 26 - 30, 2025, Chicago
Abstract #6046

Contact: Mariela Moreno Ayala (mmorenoayala@revmed.com)